Abstract No. EA60

STATENS SERUM INSTITUT

A standard protocol for the murine pneumonia model to evaluate treatments for AMR lung infections

Jon Ulf Hansen[1], Rakel Arrazuria[2], Bernhard Kerscher[2], Jennifer L. Hoover[3], Carina Vingsbo Lundberg[1], Sylvie Sordello[4], Vincent Aranzana-Climent[5], Isabelle Bekeredjian-Ding[2,6], Diarmaid Hughes[7], Philip Gribbon[8], Lena E. Friberg[5]

[1] Bacteria, Parasites & Fungi, Statens Serum Institut, Copenhagen, Denmark. [2] Division of Microbiology, Paul-Ehrlich-Institut, Langen, Germany. [3] Infectious Diseases Research Unit, GSK, Collegeville, Pennsylvania, USA. [4] Infectious Diseases, Evotec, Toulouse, France. [5] Department of Pharmacy, Uppsala University, Uppsala, Sweden. [6] Institute of Medical Microbiology, Immunology and Parasitology, University Hospital Bonn, Bonn, Germany, [7] Department of Medical Biochemistry and Microbiology, Uppsala University, Uppsala, Sweden. [8] Fraunhofer Institute for Translational Medicine and Pharmacology ITMP, Discovery Research ScreeningPort, Hamburg, Germany.

Poster contact: JOA@SSI.dk

BACKGROUND & METHOD

Preclinical *in vivo* PK/PD models play a crucial role in antimicrobial efficacy investigations and provide the basis for the selection of dosing regimens in clinical applications. Differences in the methodology in the preclinical *in vivo* models used are extensive, thus limiting the results' comparability and reproducibility and possibly impeding successful translation to the clinic. To facilitate translation, and to accelerate the development of new antibiotics, the COMBINE consortium aims to establish a reliable and globally harmonized preclinical *in vivo* mouse pneumonia model. Supported by a systematic literature review, we identified important experimental variables and discussed the relevance of these parameters through a workshop with experts in the field. Using a small panel of reference and contemporary clinical *Pseudomonas aeruginosa* and *Klebsiella pneumoniae* isolates we report a first test of the application of the standard mouse lung infection protocol.

Figure 1

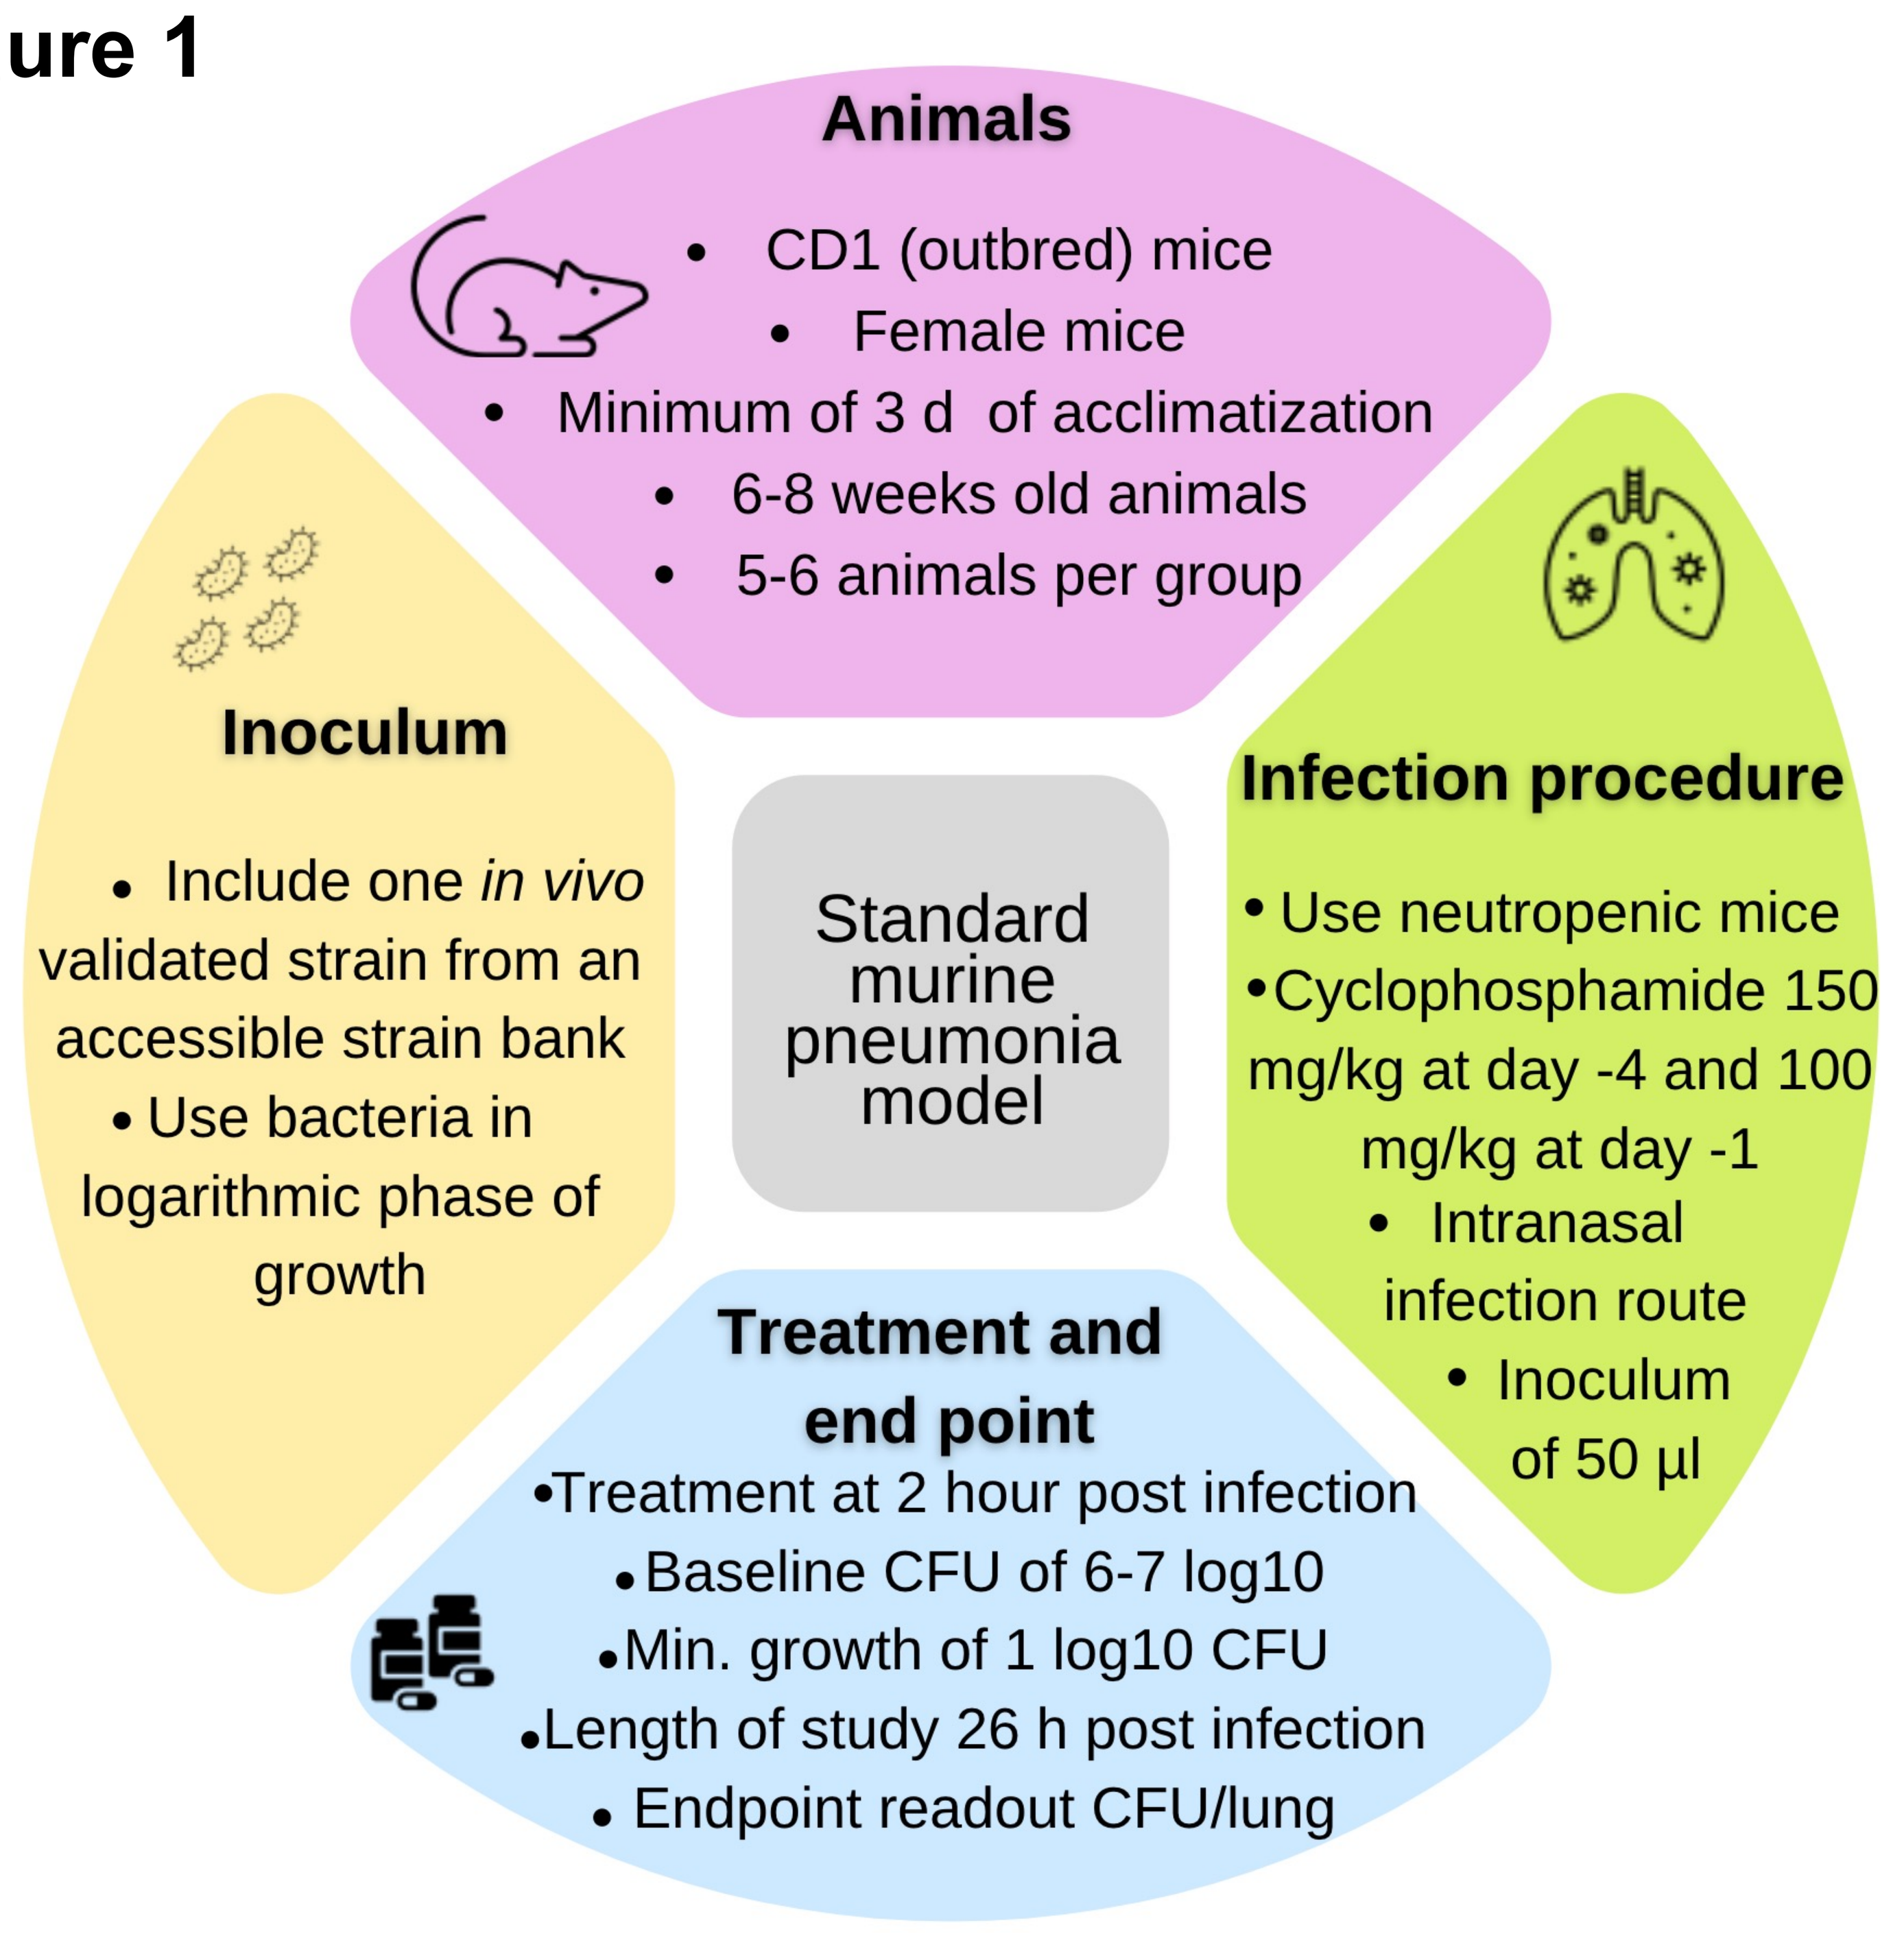

Animals
• CD1 (outbred) mice
• Female mice
• Minimum of 3 d of acclimatization
• 6-8 weeks old animals
• 5-6 animals per group
Inoculum
• Include one *in vivo* validated strain from an accessible strain bank
• Use bacteria in logarithmic phase of growth
Standard murine pneumonia model
Infection procedure
• Use neutropenic mice
• Cyclophosphamide 150 mg/kg at day -4 and 100 mg/kg at day -1
• Intranasal infection route
• Inoculum of 50 µl
Treatment and end point
• Treatment at 2 hour post infection
• Baseline CFU of 6-7 log10
• Min. growth of 1 log10 CFU
• Length of study 26 h post infection
• Endpoint readout CFU/lung

Figure 2

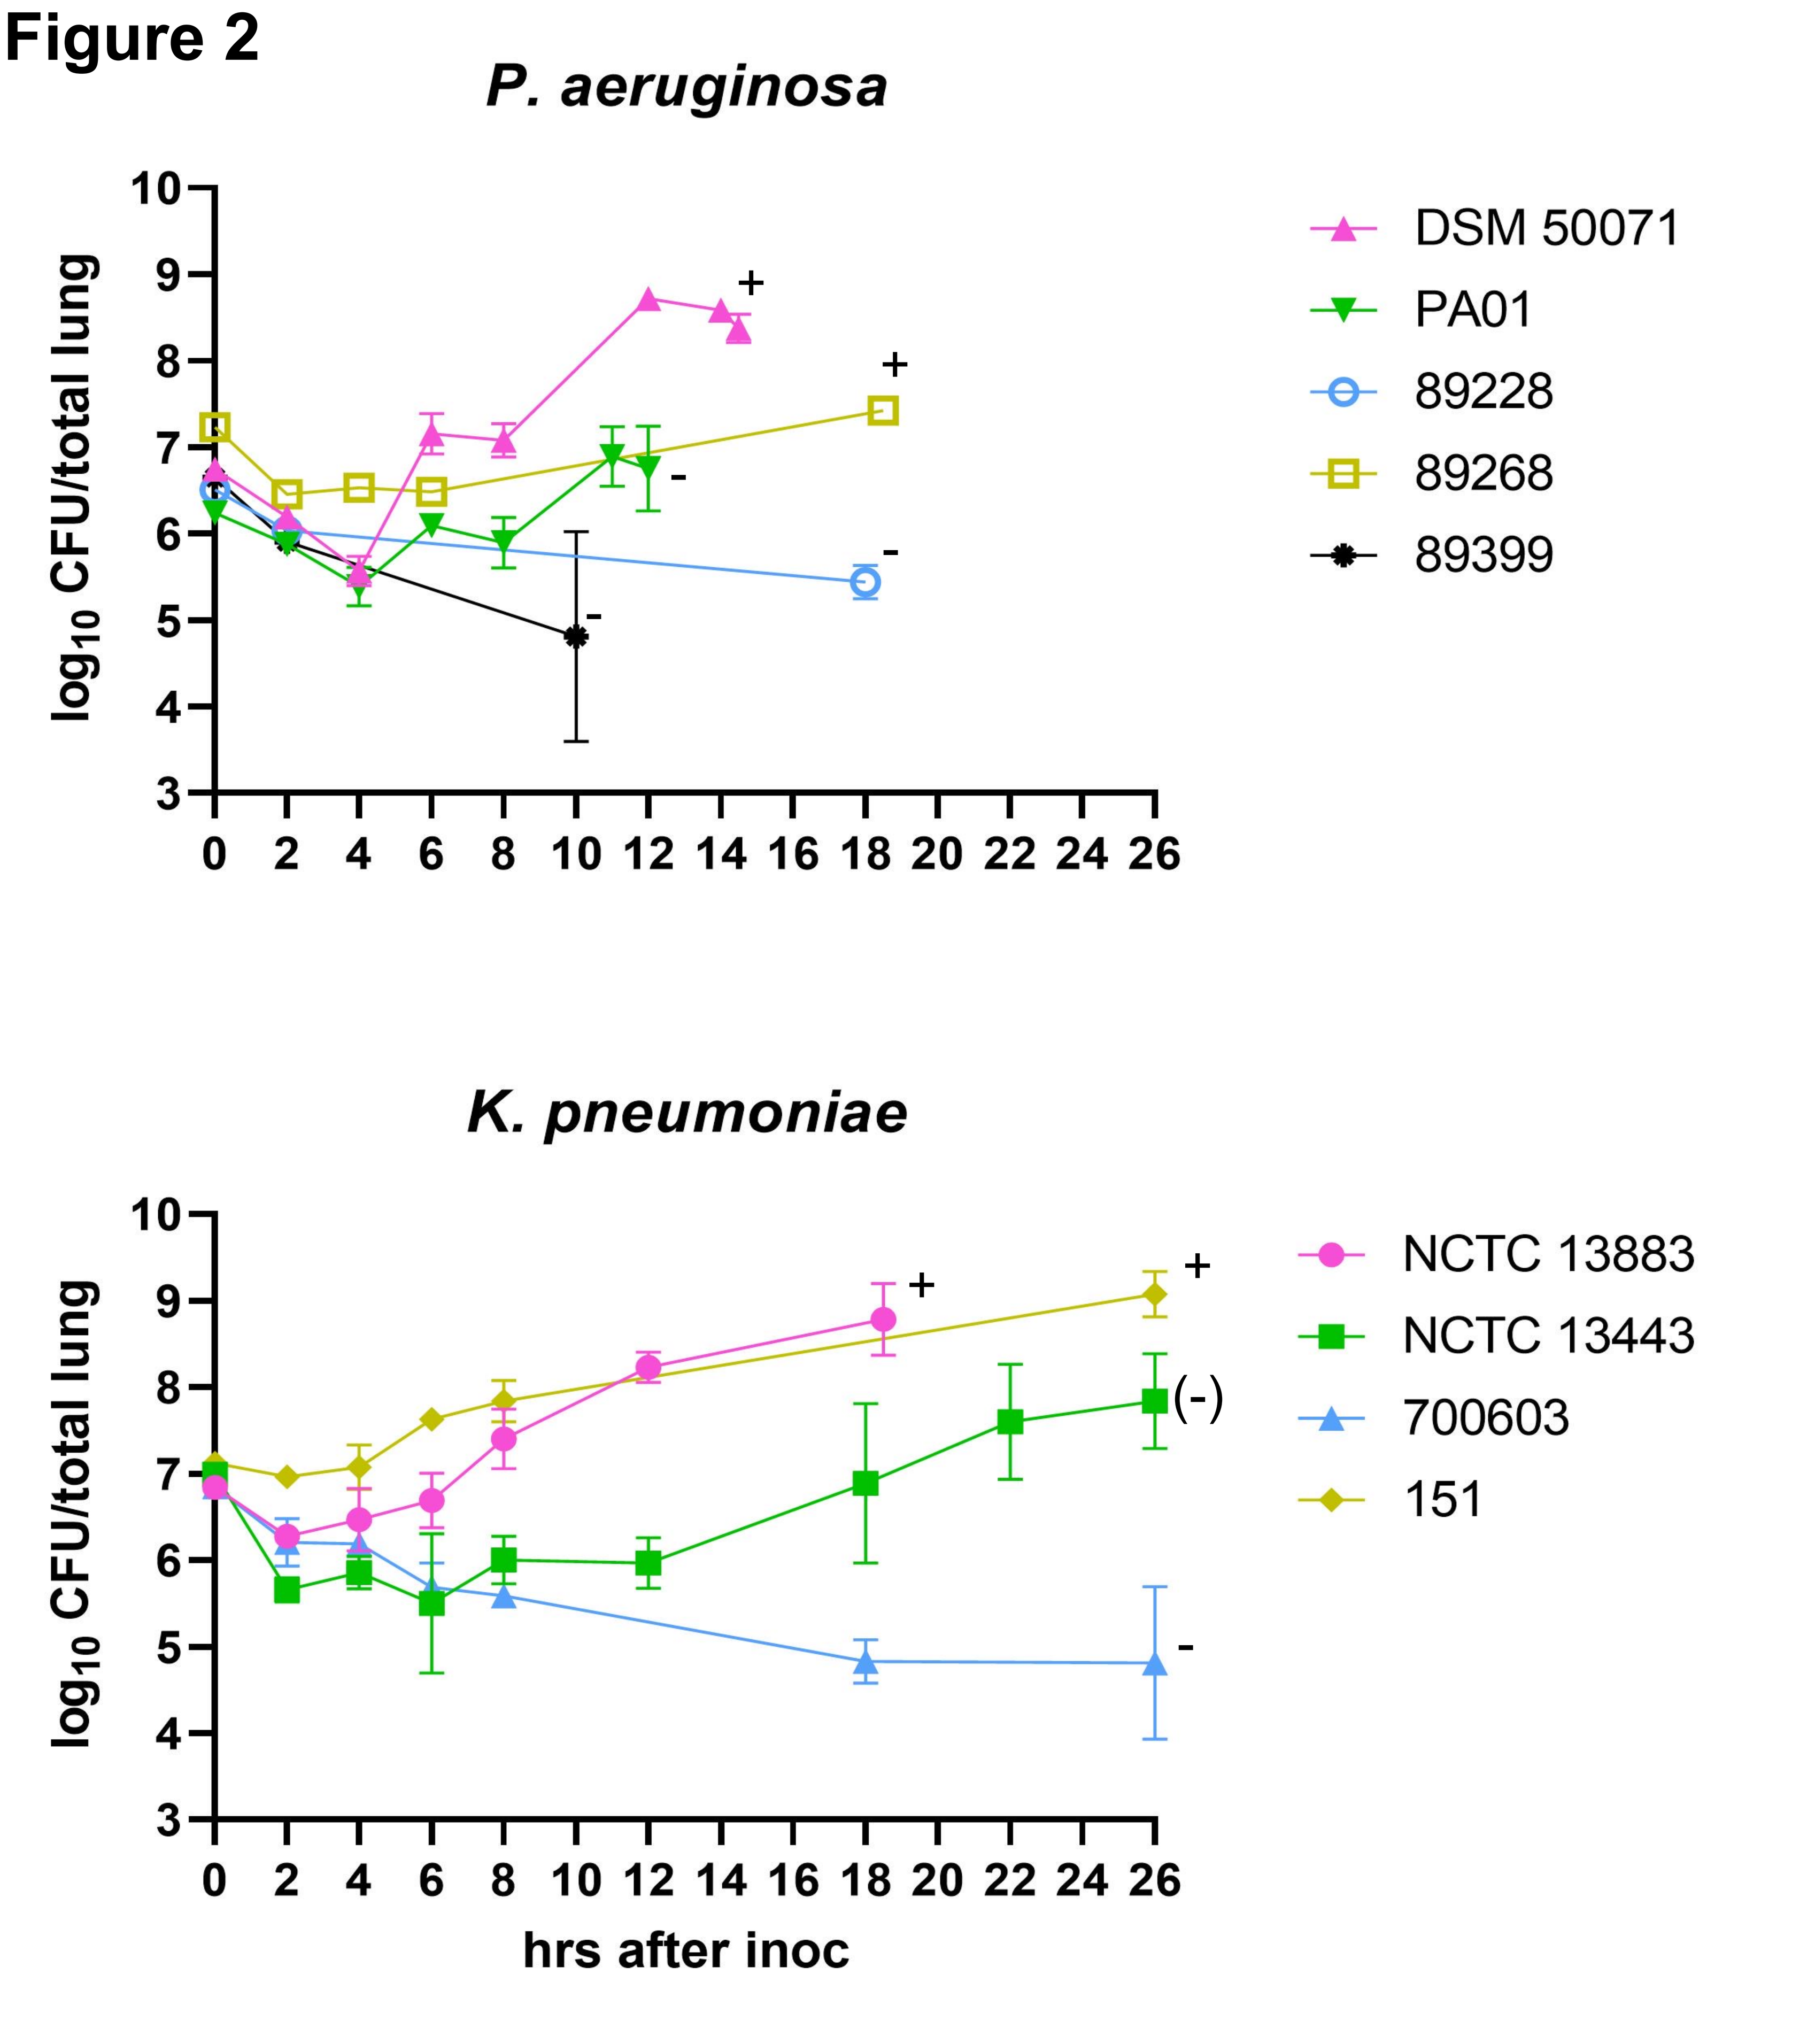

P. aeruginosa
log10 CFU/total lung
DSM 50071
PA01
89228
89268
89399
K. pneumoniae
log10 CFU/total lung
hrs after inoc
NCTC 13883
NCTC 13443
700603
151

Figure 2. Mean ± SD CFU values of examples of *P. aeruginosa* and *K. pneumoniae* isolates that meet (+) or fail (-) the criteria of the model. CFU values at t=0 were quantified from the inoculum, not mouse lungs; isolates with endpoints before 26 hrs were euthanized due to clinical score

Table 1

Species	No. of isolates tested	Increase in bact. load < 1 log from 2 hrs to endpoint	Lethal ≤12hrs	Suitable strains identified
P. aeruginosa	12	2	8	2
K. pneumoniae	11	2	0	9

RESULTS

A standard mouse pneumonia protocol was established (figure 1) and applied to test susceptible as well as AMR isolates of species *K. pneumoniae* and *P. aeruginosa* isolates (table 1), to identify strains that meet the criteria for use in the COMBINE standard protocol (exemplified in figure 2). Overall, the majority of the tested *K. pneumoniae* strains met the established virulence criteria whereas the majority of *P. aeruginosa* strains were either lethal in less than 12 hrs or did not grow in vivo. Ultimately, a selection of isolates that perform appropriately in the model, together with PK/PD data for reference antibiotics vs. these isolates, will be made available to the community for bench-marking new small molecule antibiotics in preclinical development.

We thank the GNA NOW project and in particular Sharon Evans and Pippa Griffin from the North Bristol NHS Trust for kindly sharing clinical *P. aeruginosa* and *K. pneumoniae* isolates. Also Frederikke Rosenborg, Karen Juhl, Sandra N.B. Jensen and Tina S. Lundager (Statens Serum Institut) for expert technical assistance. All animal studies were ethically reviewed and carried out in accordance with European Directive 2010/63/EEC and the GSK Policy on the Care, Welfare and Treatment of Animals.

This project has received funding from the Innovative Medicines Initiative 2 Joint Undertaking under grant agreement No 853967. This Joint Undertaking receives support from the European Union's Horizon 2020 research and innovation programme and EFPIA companies' in kind contribution. This communication reflects the views of authors from the COMBINE consortium and neither IMI nor European Union and EFPIA are liable for any use that may be made of the information herein.
COMBINE - IMI AMR Accelerator (amr-accelerator.eu)

efpia

innovative medicines initiative

COMBINE Part of IMI AMR Accelerator