A Standard Protocol for the Murine Pneumonia Model to Investigate Antibiotic Treatment Effect of MDR Bacteria Infections

Poster contact: CVL@SSI.DK

Carina Vingsbo Lundberg[1], Jon Ulf Hansen[1], Diego Vera[2], Rakel Arrazuria[3], Bernhard Kerscher[3], Jennifer L. Hoover[4], Sylvie Sordello[5], Isabelle Bekeredjian-Ding[3,6], Diarmaid Hughes[7], Lena E. Friberg[2]

[1] Bacteria, Parasites & Fungi, Statens Serum Institut, Copenhagen, Denmark. [2] Dept. of Pharmacy, Uppsala University, Uppsala, Sweden [3] Div. of Microbiology, Paul-Ehrlich-Institut, Langen, Germany. [4] Infectious Diseases Research Unit, GSK, Collegeville, Pennsylvania, USA. [5] Infectious Diseases, Evotec, Toulouse, France. [6] Institute of Medical Microbiology, Immunology and Parasitology, University Hospital Bonn, Bonn, Germany, [7] Dept. of Medical Biochemistry and Microbiology, Uppsala University, Uppsala, Sweden.

Introduction

Preclinical *in vivo* PK/PD models play a crucial role in antimicrobial efficacy investigations. Differences in the methodology in the models used are extensive and may limit the comparability, reproducibility and translation to the clinic. To support the development of new antibiotics, the COMBINE consortium has established a standard protocol for acute murine gram-negative pneumonia and initiated the characterization of susceptible as well as multi-drug resistant isolates of *K. pneumoniae* and *P. aeruginosa* isolates.

Table 1	Statens Serum Institut (SSI)	Paul-Ehrlich-Institut (PEI)
Animals (vendor)	Hsd:ICR (ENVIGO)	RjOrl:SWISS (JANVIER)
Inoculum growth	5% horse blood agar plates	Trypticase soy broth
Anesthesia	General anesthesia (parenteral): Zolazepam 15 mg/kg; Tiletamine 15 mg/kg; Xylazine 24 mg/kg; Butorphanol: 0.3 mg/kg	Isoflurane (inhalation)
Age (weeks)	7-8	6-7
Lung processing for CFU determination	Stored at -80 ˚C	Freshly processed
Culture media for CFU determination	Selective media: blue (bromothymol) agar / Cetrimide agar	Non selective media: Trypticase soy agar

Methods: The COMBINE protocol

Important experimental variables were discussed and the relevance confirmed through a workshop with experts in the field (ref 1, 2) and subsequently a standard protocol was suggested (Fig 1).

The reproducibility of the virulence of 10 selected isolates, using the standard protocol was evaluated at two different laboratories, SSI and PEI. The protocol contains a few non-standardized variables or parameters because of different institution practices. Differences between the SSI and PEI protocols are shown in table 1.

Fig 1.

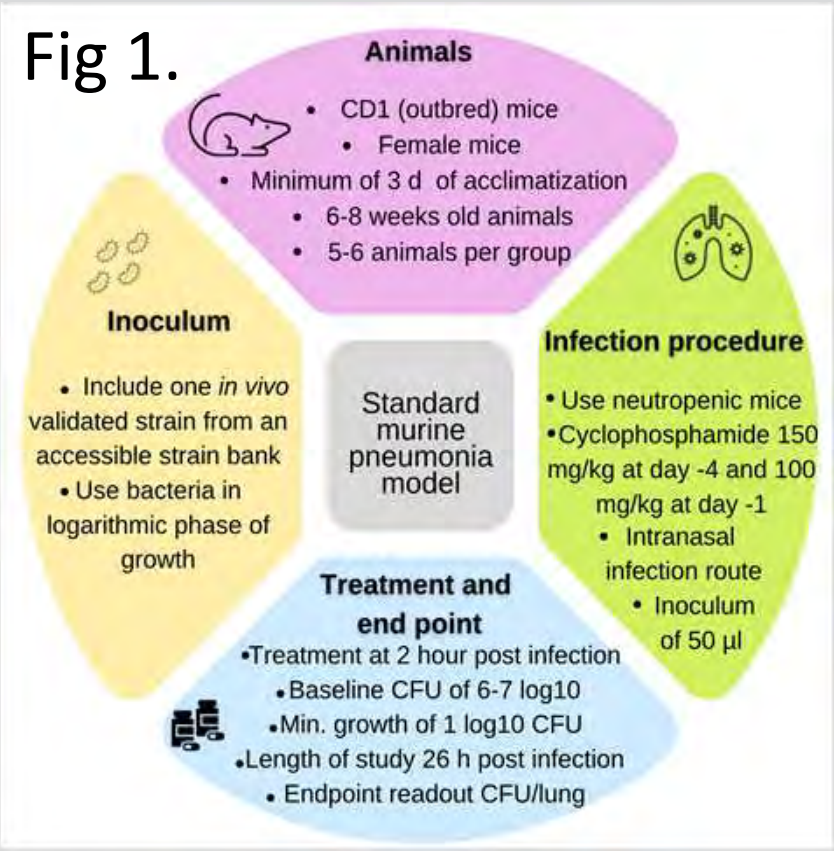
Animals
• CD1 (outbred) mice
• Female mice
• Minimum of 3 d of acclimatization
• 6-8 weeks old animals
• 5-6 animals per group
Inoculum
• Include one in vivo validated strain from an accessible strain bank
• Use bacteria in logarithmic phase of growth
Standard murine pneumonia model
Infection procedure
• Use neutropenic mice
• Cyclophosphamide 150 mg/kg at day -4 and 100 mg/kg at day -1
• Intranasal infection route
• Inoculum of 50 µl
Treatment and end point
• Treatment at 2 hour post infection
• Baseline CFU of 6-7 log10
• Min. growth of 1 log10 CFU
• Length of study 26 h post infection
• Endpoint readout CFU/lung

Results: Virulence and reproducibility in the COMBINE standard protocol

Virulence evaluation of bacterial isolates in the COMBINE standard protocol

Out of 33 tested isolates, 15 isolates showed a suitable virulence applying the standard protocol at Statens Serum Institut. The virulence was confirmed in 2-3 studies for each isolate. Virulence criteria for strain selection was defined as a minimum of 1 $\log_{10}$ bacterial growth in the lungs and survival of mice for ≥12 hours post inoculation. These 15 isolates have been made available to the scientific community in a biorepository at DSMZ (ref. 3)

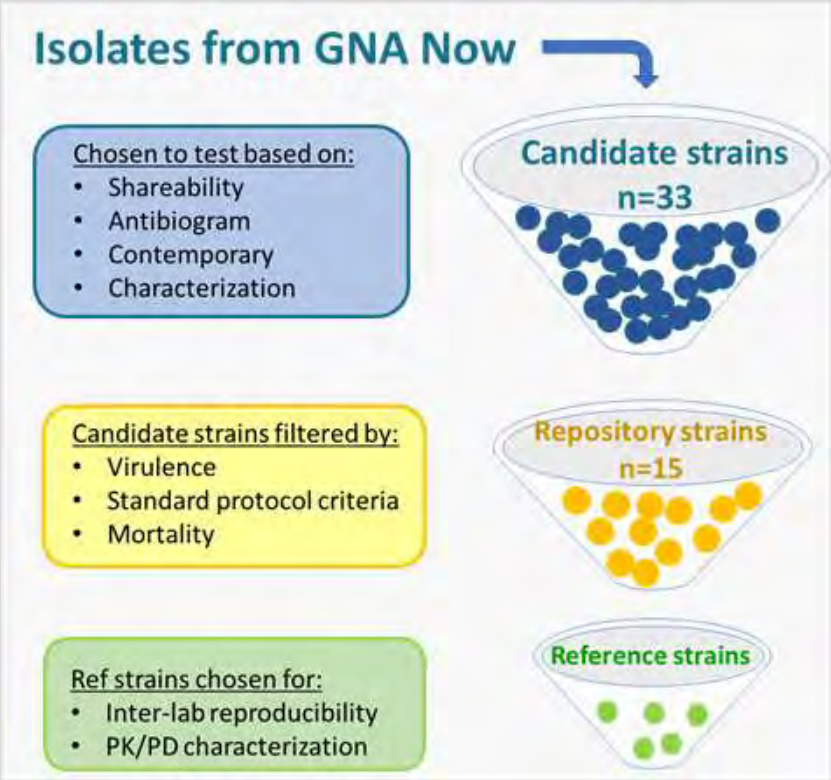
Isolates from GNA Now
Chosen to test based on:
• Shareability
• Antibiogram
• Contemporary
• Characterization
Candidate strains n=33
Candidate strains filtered by:
• Virulence
• Standard protocol criteria
• Mortality
Repository strains n=15
Ref strains chosen for:
• Inter-lab reproducibility
• PK/PD characterization
Reference strains

Fig 2 Characterization and selection process of strains for the standard model

Assessment of the reproducibility of the COMBINE standard protocol

10 isolates were selected for confirmatory virulence studies at the Paul-Ehrlich-Institut. The use of the COMBINE standard protocol led to good reproducibility of virulence at both laboratories. Examples of in vivo growth curves with 4 of these isolates are shown in figure 3.

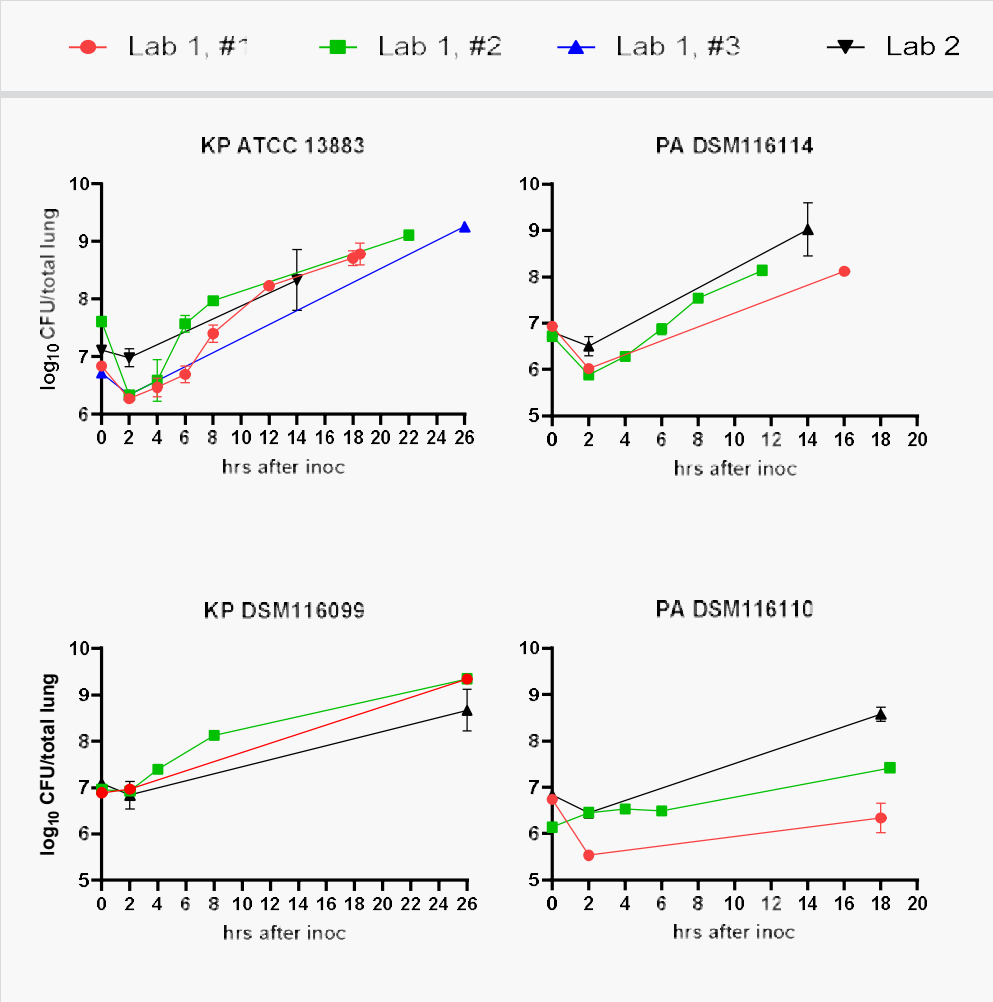
Lab 1, #1
Lab 1, #2
Lab 1, #3
Lab 2
KP ATCC 13883
PA DSM116114
KP DSM116099
PA DSM116110
log10 CFU/total lung
hrs after inoc

Fig 3 Reproducibility of the virulence of strains in the standard model

Adaptation of the standard protocol for the use with MRSA

The standard protocol was applied to methicillin resistant *Staphylococcus aureus* (MRSA). Clinical isolates from the respiratory tract (SSI strain collection) were selected and tested at SSI for virulence in the standard protocol. None of the 8 isolates tested met the virulence criteria. By switching to Balb/C mice, one isolate met the virulence criteria of 1 log growth in the lungs and survival of mice for ≥12 hours post inoculation (figure 4)

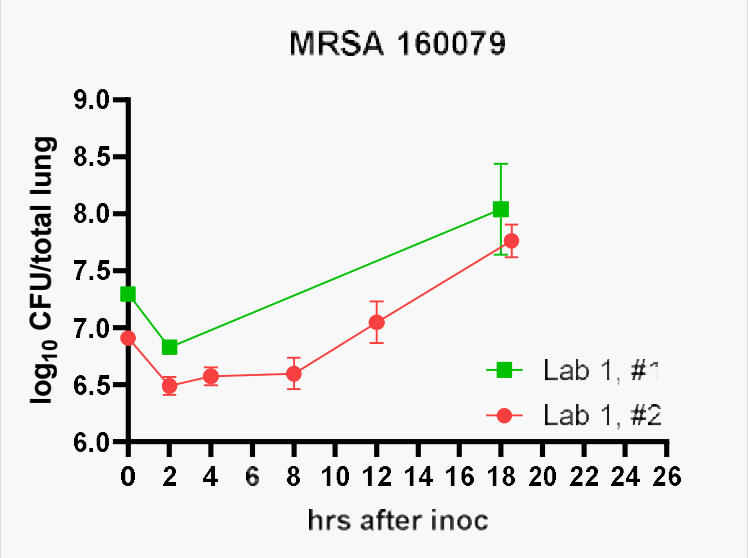
MRSA 160079
log10 CFU/total lung
hrs after inoc
Lab 1, #1
Lab 1, #2

Fig 4 Performance of MRSA in the standard model

Conclusion

- We confirm that the virulence of 10 reference strains is reproducible across laboratories despite a slight variation of a few non-standardized variables in the standard protocol.
- Modification of the standard protocol was used to establish an MRSA pneumonia model.

Future Perspective

The robustness of the standard model is being further evaluated in PK/PD studies with reference antibiotics. The generated PK/PD data, will be made available to the community for benchmarking new small molecule antibiotics in preclinical development.

We thank the GNA NOW project and in particular Marie Attwood, Sharon Evans and Pippa Griffin from the North Bristol NHS Trust for kindly sharing clinical *P. aeruginosa* and *K. pneumoniae* isolates. Also Frederikke Rosenborg, Karen Juhl, Sandra Jensen and Tina Lundager (Statens Serum Institut) for expert technical assistance.

This factsheet reflects the authors' view and neither IHI nor the European Union, EFPIA, or any Associated Partners are responsible for any use that may be made of the information contained herein.

All animal studies were ethically reviewed and carried out in accordance with European Directive 2010/63/EEC and the GSK Policy on the Care, Welfare and Treatment of Animals.

References

1. Rakel Arrazuria et.al. Variability of murine bacterial pneumonia models used to evaluate antimicrobial agents. Front. Microbiol. 2022
2. Rakel Arrazuria et.al. Expert Workshop Summary: Advancing towards a standardized murine model to evaluate treatments for AMR lung infections. Front. Microbiol. 2022
3. COMBINE Preclinical Bacterial Strain Repository https://www.pei.de/EN/regulation/reference-material/reference-material-node

www.imi.europa.eu

This project has received funding from the Innovative Medicines Initiative 2 Joint Undertaking under Grant Agreement No 853967. This Joint Undertaking receives support from the European Union's Horizon 2020 research and innovation programme and EFPIA companies' in kind contribution.